TUPE0068

HLA-B*5701 Screening Reduces Abacavir Hypersensitivity Reactions (ABC HSR); Multiple Lines of Evidence to Establish a New Standard of Care

Wannamaker P[1], Mallal S[2], Phillips E[2], Young B[3], Brothers C[1], Sutherland-Phillips D[1], Thorborn D[4], and Shaefer M[1]

[1]GlaxoSmithKline Research and Development, Research Triangle Park, NC, USA; [2]Murdoch University, Perth, Australia;
[3]Denver ID Associates, Denver, CO, USA; [4]GlaxoSmithKline Pharma Europe, Brentford, UK

Abstract

Background: ABC HSR has been associated with the pharmacogenetic marker HLA-B*5701 in several studies. However, evidence of the clinical utility of screening is required from controlled clinical trials, open screening programs and from different target populations to justify uptake into clinical practice.

Methods: PREDICT-1 was a prospective, randomized, controlled, double-blinded study enrolling European and Australian subjects to compare the incidence of ABC HSR with or without prospective HLA-B*5701 screening. SHAPE was a retrospective case-control study to estimate the sensitivity of HLA-B*5701 in Black and White subjects in the United States with ABC HSR. ARIES is a prospective treatment simplification study of ABC/3TC + atazanavir/ritonavir in *HLA-B*5701* negative ART-naïve subjects across the United States and Canada. Skin patch testing (SPT) was used to provide immunologic confirmation of suspected ABC HSR in all studies.

Results: In the predominantly white population enrolled in PREDICT-1, 5.6% of subjects carried HLA-B*5701. SPT-positive cases of ABC HSR were reduced from 2.7% to 0% in the screening arm (7.8% to 3.4%, all cases) (p<0.001). SHAPE demonstrated a 100% sensitivity of *HLA-B*5701* for SPT-positive ABC HSR in both White (n=42) and Black (n=5) subjects. In ARIES, which employed prospective HLA-B*5701 screening, 725 ART-naïve subjects were screened of which 32 (7.2%) of Whites and 7 (2.8%) Blacks were positive for *HLA-B*5701* and excluded. After open screening and initiation of ABC, suspected HSR was reported in 4 subjects (0.8%) through 18 weeks, none were SPT-positive.

Conclusions: Results from PREDICT-1, SHAPE and ARIES provide conclusive evidence of the validity, generalizability and effectiveness of *HLA-B*5701* screening as a standard of care approach before starting an abacavir containing regimen. Although screening dramatically reduces the risk of ABC HSR, appropriate clinical management of suspected HSR remains paramount.

Introduction

- Research into the genetic basis of the abacavir hypersensitivity reaction has identified a strong association between the *HLA-B*5701* allele and clinical abacavir hypersensitivity.[1, 2]
- The use of an abacavir skin-patch test has helped to improve the diagnostic precision of ABC HSR in research settings.[3]
- An immunogenetic basis for *HLA-B*5701*-restricted ABC HSR has been identified, further strengthening this correlation.[4]
- Given the strong relationship of this pharmacogenetic marker for ABC HSR and the widespread availability of this test, the clinical utility of this marker can be demonstrated through several lines of research, including:
 - Results from a large, prospective, blinded, randomized study demonstrating significant benefits in risk reduction for developing ABC HSR with the use of *HLA-B*5701* screening.
 - Compelling data suggesting the generalizability of screening across diverse populations.
 - Large prospective open screening studies consistently demonstrating significant reduction in rates of suspected ABC HSR post-screening.
 - Results from "real-world" populations demonstrating efficient and effective screening practices.

Methods

- GSK undertook three studies to address the clinical utility of *HLA-B*5701* screening to reduce the incidence of ABC HSR. Study designs are below:

Figure 1. PREDICT-1 Study Design

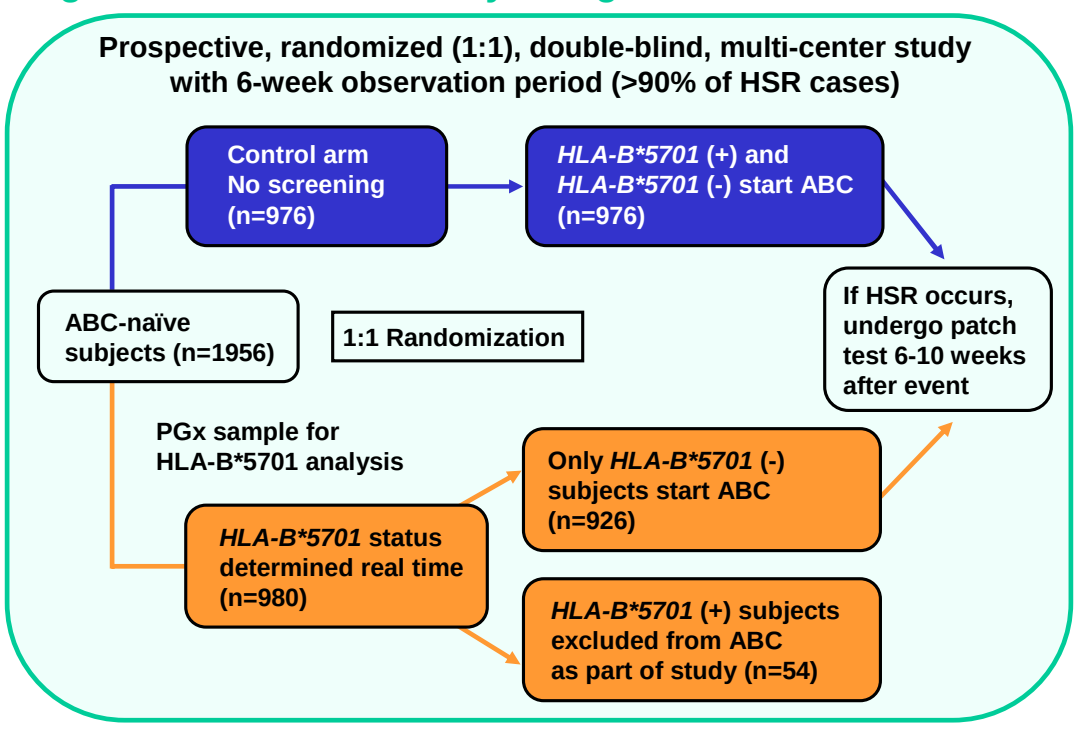
Prospective, randomized (1:1), double-blind, multi-center study with 6-week observation period (>90% of HSR cases)
Control arm No screening (n=976)
HLA-B*5701 (+) and HLA-B*5701 (-) start ABC (n=976)
ABC-naïve subjects (n=1956)
1:1 Randomization
If HSR occurs, undergo patch test 6-10 weeks after event
PGx sample for HLA-B*5701 analysis
Only HLA-B*5701 (-) subjects start ABC (n=926)
HLA-B*5701 status determined real time (n=980)
HLA-B*5701 (+) subjects excluded from ABC as part of study (n=54)

Figure 2. SHAPE Study Design

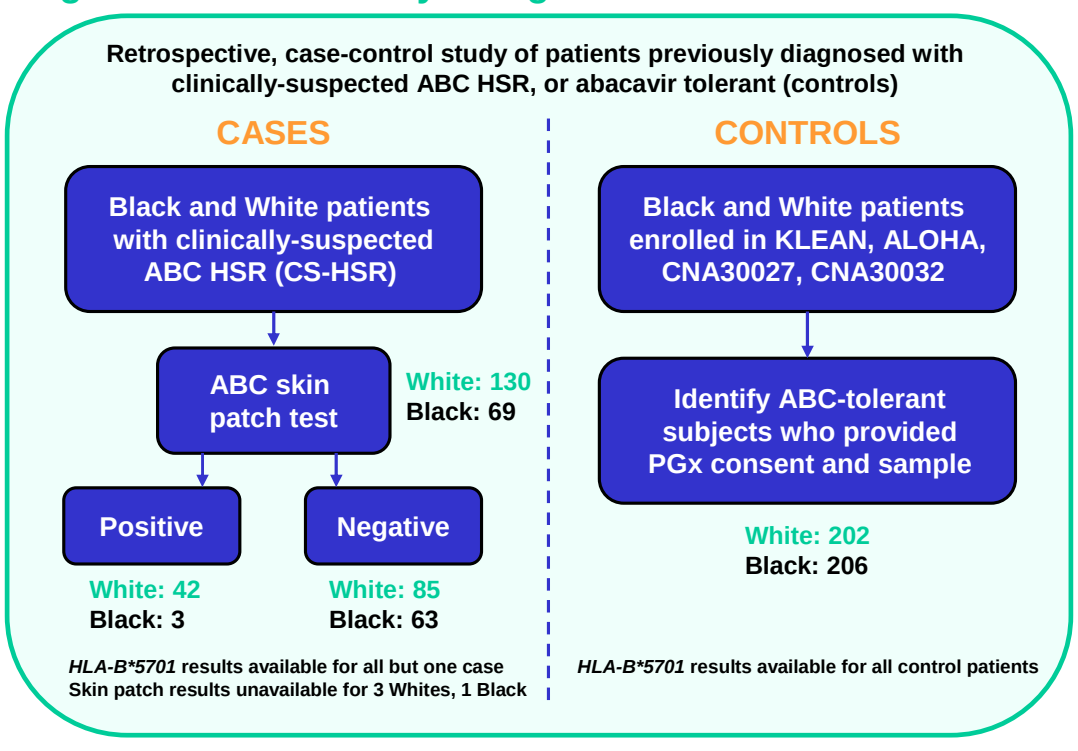
Retrospective, case-control study of patients previously diagnosed with clinically-suspected ABC HSR, or abacavir tolerant (controls)
CASES
CONTROLS
Black and White patients with clinically-suspected ABC HSR (CS-HSR)
Black and White patients enrolled in KLEAN, ALOHA, CNA30027, CNA30032
ABC skin patch test
White: 130
Black: 69
Identify ABC-tolerant subjects who provided PGx consent and sample
Positive
Negative
White: 42
Black: 3
White: 85
Black: 63
White: 202
Black: 206
HLA-B*5701 results available for all but one case
Skin patch results unavailable for 3 Whites, 1 Black
HLA-B*5701 results available for all control patients

Figure 3. ARIES Study

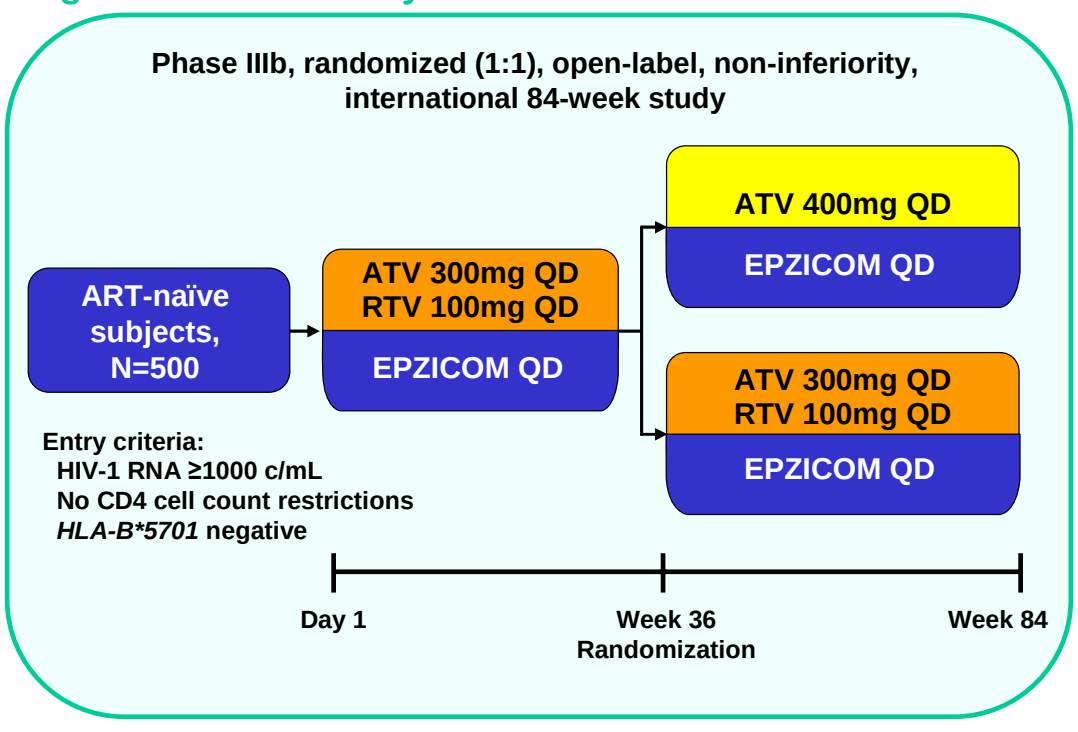
Phase IIIb, randomized (1:1), open-label, non-inferiority, international 84-week study
ART-naïve subjects, N=500
ATV 300mg QD
RTV 100mg QD
EPZICOM QD
ATV 400mg QD
EPZICOM QD
ATV 300mg QD
RTV 100mg QD
EPZICOM QD
Entry criteria:
HIV-1 RNA ≥1000 c/mL
No CD4 cell count restrictions
HLA-B*5701 negative
Day 1
Week 36
Randomization
Week 84

Results

The PREDICT-1 study demonstrated a statistically significant reduction in the incidence of clinically-suspected ABC HSR (CS-HSR) after exclusion of subjects who carried *HLA-B*5701,* and none of these subjects had immunologically-confirmed ABC HSR (IC-HSR) by skin patch testing (Figure 4).[5]

Figure 4. PREDICT-1 Study Results

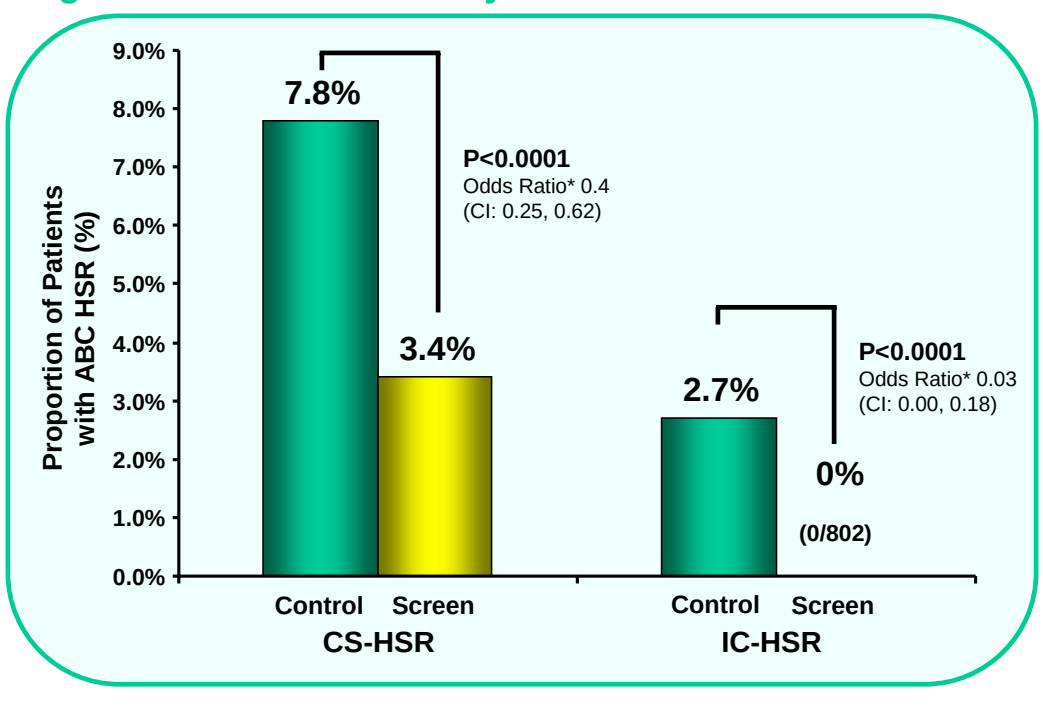
9.0%
8.0%
7.0%
6.0%
5.0%
4.0%
3.0%
2.0%
1.0%
0.0%
Proportion of Patients with ABC HSR (%)
7.8%
3.4%
P<0.0001
Odds Ratio* 0.4
(CI: 0.25, 0.62)
2.7%
0%
(0/802)
P<0.0001
Odds Ratio* 0.03
(CI: 0.00, 0.18)
Control
Screen
CS-HSR
Control
Screen
IC-HSR

The SHAPE study demonstrated a high (100%) sensitivity of *HLA-B*5701* in both White and Black subjects who had an ABC HSR that was immunologically-confirmed by a positive ABC skin patch test. Specificity of *HLA-B*5701* in a group of ABC-tolerant white and black controls was 96% and 99%, respectively (Figure 5).[6]

Figure 5. SHAPE Study Results

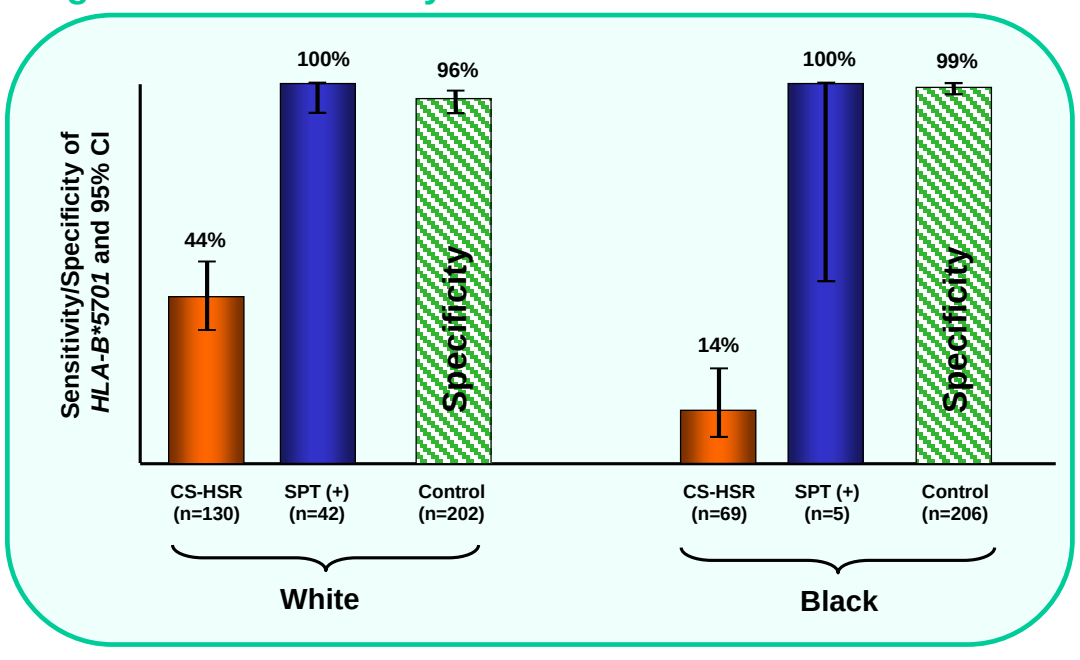
Sensitivity/Specificity of HLA-B*5701 and 95% CI
44%
100%
96%
Specificity
14%
100%
99%
Specificity
CS-HSR (n=130)
SPT (+) (n=42)
Control (n=202)
CS-HSR (n=69)
SPT (+) (n=5)
Control (n=206)
White
Black

In ARIES, 725 subjects were screened for entry; 41 (5.7%) were found to be *HLA-B*5701* positive and were excluded. These included 32 (7.2%) Whites, 7 (2.8%) Black and 2 (5.6%) Other (American Indian [1], Asian [1]) race.

The ARIES study demonstrated that in a diverse US population (34% Black), suspected ABC HSR was rarely reported (4/ 517, 0.8%) after open-label *HLA-B*5701* screening through 36 weeks, and that none of these cases were immunologically-confirmed by ABC skin patch test. This result is dramatically lower than recent similar studies where *HLA-B*5701* screening was not used. (Figure 6).[7]

Figure 6. ARIES ABC HSR Results vs. Other Recent Studies*

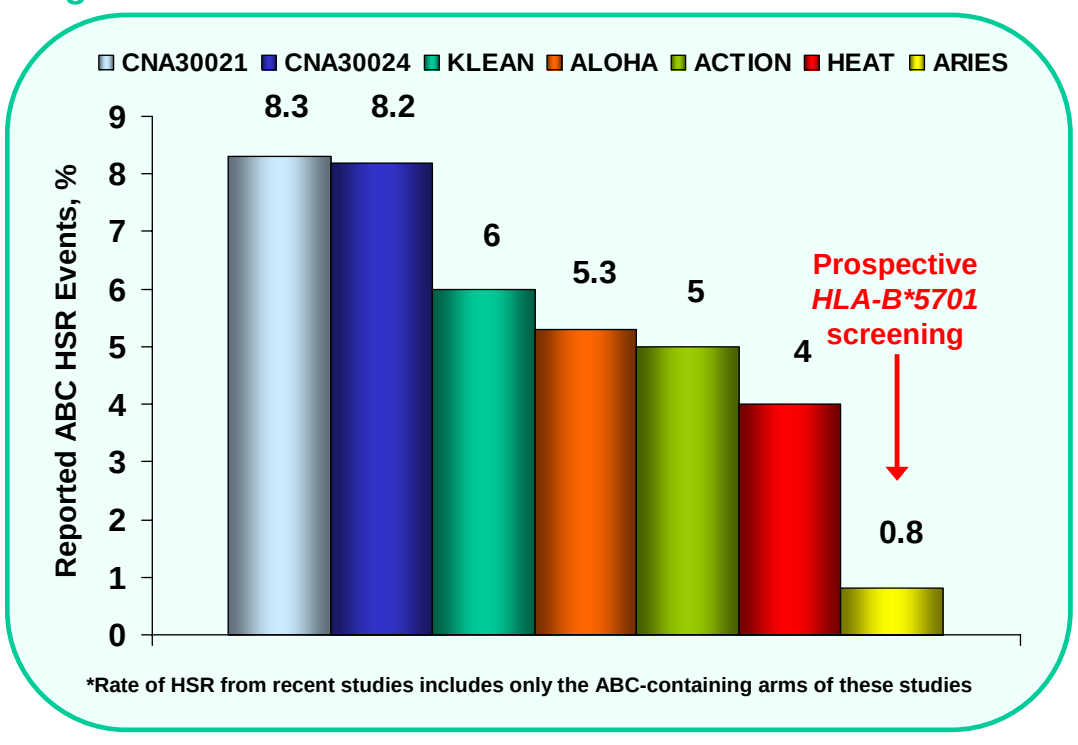
CNA30021
CNA30024
KLEAN
ALOHA
ACTION
HEAT
ARIES
Reported ABC HSR Events, %
9
8
7
6
5
4
3
2
1
0
8.3
8.2
6
5.3
5
4
0.8
Prospective HLA-B*5701 screening

*Rate of HSR from recent studies includes only the ABC-containing arms of these studies

Independent cohorts in France, Australia and the UK have also reported similar reductions in the rate of suspected ABC HSR after initiation of an *HLA-B*5701* screening program prior to initiation of abacavir therapy.[8, 9, 10]

Discussion

- The PREDICT-1 study used a robust, randomized, prospective double-blinded study design to substantiate the clinical benefit of *HLA-B*5701* screening to reduce ABC HSR events in a predominantly white population; however, its blinded study design precluded the ability to directly realize the impact that prospective knowledge of *HLA-B*5701* status has on reduction in false positive clinical diagnosis.
- The SHAPE study, using a case-control design, demonstrated the high sensitivity of *HLA-B*5701* for ABC HSR to be generalizable across White and Black racial groups, representing the major target populations with the highest and lowest allele frequencies, respectively, and supporting generalizability of the utility of *HLA-B*5701* screening across race.
- In a large and diverse US population, the ongoing ARIES study resulted in a dramatic reduction in ABC HSR rates compared to similar historical studies.
 - The results from ARIES are fully consistent with both the PREDICT-1 and SHAPE studies, as well as multiple independent open screening cohorts, demonstrating a marked reduction in all suspected ABC HSR events, none of which were confirmed by ABC skin patch testing.
 - The open-label design of the ARIES study is a further evolution of the *HLA-B-5701* screening concept which reflects clinical practice, where a diverse ABC-naïve population was treated prospectively and screening results were known both by patients and prescribers.
- This pharmacogenetics research program, from the initial identification of the association of *HLA-B*5701* with risk of ABC HSR, independent replication of the association, and demonstration of the clinical utility of *HLA-B*5701* screening on patient safety, provides a model for other researchers toward the incorporation of PGx and other biomarkers into clinical practice.

Conclusions

- Screening for *HLA-B*5701* and avoidance of ABC in those who are positive for the allele reduces the risk of abacavir hypersensitivity reaction.
 - The PREDICT-1, SHAPE and ARIES studies provide the evidence of validity, generalizability and effectiveness to establish screening as a standard of care approach where available.
- While *HLA-B*5701* screening is an effective tool to reduce ABC HSR, appropriate clinical management of suspected HSR remains critical.

Acknowledgements

The authors would like to acknowledge the many patients, investigators, researchers and others whose work has made this effort possible.

References

[1]Mallal S, Nolan D, Witt C, *et. al*. Association between presence of *HLA-B*5701*, *HLA-DR7*, and *HLA-DQ3* and hypersensitivity of HIV-1 reverse transcriptase inhibitor abacavir. *Lancet* 2002; 359:727-732.

[2]Hetherington S, Hughes A, Mosteller M, *et. al*. Genetic variations in HLA-B region and hypersensitivity reactions to abacavir. *Lancet* 2002; 359:1121-1122.

[3]Philllips E, Sulivan J, Knowles S, *et. al*. Utility of patch testing in patients with hypersensitivity syndromes associated with abacavir. *AIDS* 2002; 16:2223-2225.

[4]Chessman D, Kostenko L, Lethborg T, *et. al*. Human leukocyte antigen class 1-restricted activation of $CD8^+$ T cells provides the immunogenetic basis of a systemic drug hypersensitivity. *Immunity* 2008; 28:822-832.

[5]Mallal S, Phillips E, Carosi G, *et. al.* HLA-B*5701 screening for hypersensitivity to abacavir. *N Engl. J. Med*. 2008; 358:568-579.

[6]Saag M, Balu R, Phillips E, *et. al*. High sensitivity of human leukocyte antigen-B*5701 as a marker for immunologically confirmed abacavir hypersensitivity in white and black patients. *CID* 2008;46: 1111-1118.

[7]Young B, Squires K, Patel P, et al. First large, multicenter, open-label study utilizing *HLA-B*5701* screening for abacavir hypersensitivity in North America. *AIDS*, 2008; In Press.

[8]Zucman D, de Truchis, P, Majerholc C, *et al*. Prospective screening for human leukocyte antigen-B*5701 avoids abacavir hypersensitivity reaction in the ethnically mixed french HIV population. *J Acquir Immune Defic Syndr* 2007;45:1-3.

[9]Rauch A, Nolan D, Martin E, *et al*. Prospective genetic screening decreases the incidence of abacavir hypersensitivity reactions in the Western Australian HIV cohort study. *CID* 2006;43:99-102.

[10]Waters L, Mandalia S, Gazzard B, *et. al*. Prospective *HLA-B*5701* screening and abacavir hypersensitivity: a single centre experience. *AIDS* 2007;21:2533-2534.

AIDS 2008 – 17th International AIDS Conference, 3–8 August 2008, México City, México